Improvement in Liver Histology Is Observed in Most Patients With Chronic Hepatitis Delta After 48 Weeks of Bulevirtide Monotherapy

WED-392

Pietro Lampertico[1,2], Soo Aleman[3], Pavel Bogomolov[4], Tatyana Stepanova[5], Markus Cornberg[6], Sandra Ciesek[7], Annemarie Berger[7], Dmitry Manuilov[8], Mingyang Li[8], Audrey H Lau[8], Ben L Da[8], Grace M Chee[8], Maurizia R Brunetto[9,10], Stefan Zeuzem[11], Heiner Wedemeyer[6]

[1]Division of Gastroenterology and Hepatology, Foundation IRCCS Ca' Granda Ospedale Maggiore Policlinico, Milan, Italy; [2]Department of Pathophysiology and Transplantation, CRC "A. M. and A. Migliavacca" Center for Liver Disease, University of Milan, Milan, Italy; [3]Department of Infectious Diseases, Karolinska University Hospital/Karolinska Institutet, Stockholm, Sweden; [4]M.F. Vladimirsky Moscow Regional Research and Clinical Institute, Moscow, Russian Federation; [5]LLC Clinic of Modern Medicine, Moscow, Russian Federation; [6]Clinic for Gastroenterology, Hepatology, Infectious Diseases, and Endocrinology, Hannover Medical School, Hannover, Germany; [7]Institute for Medical Virology, German Centre for Infection Research, External Partner Site Frankfurt, University Hospital, Goethe University Frankfurt am Main, Frankfurt am Main, Germany; [8]Gilead Sciences, Inc., Foster City, CA, USA; [9]Hepatology Unit, Reference Center of the Tuscany Region for Chronic Liver Disease and Cancer, University Hospital of Pisa and Department of Clinical and Experimental Medicine, University of Pisa, Pisa, Italy; [10]Department of Clinical and Experimental Medicine, University of Pisa, Pisa, Italy; [11]Department of Medicine, University Hospital Frankfurt, Frankfurt am Main, Germany

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

Conclusions

- In a subanalysis of patients with paired BL and W48 liver biopsies, an improvement in liver histology was observed in most patients with CHD after 48 weeks of BLV monotherapy, including those who did not achieve VR
- Decreases in ALT levels and/or HAI were observed in most patients treated with BLV
 - — The degrees of improvement in HAI and ALT levels did not necessarily correlate
- Histologic improvement and improvement in HAI or HAI category occurred more frequently in patients with VR or PR
- An improvement in fibrosis occurred most frequently in patients with VR
- No consistent pattern of HAI, ALT level, or fibrosis change was observed in the control group

Plain Language Summary

- An improvement in liver histology, or how liver cells look under a microscope, was observed in most patients with chronic hepatitis delta after 48 weeks of bulevirtide treatment
- This improvement in liver histology was observed even in patients who did not have a large decrease in the amount of hepatitis delta virus from bulevirtide treatment

References: 1. Stockdale AJ, et al. *J Hepatol*. 2020;73:523-32. 2. Hepcludex. European Medicines Agency SmPC. Gilead Sciences, Inc.; 2023. 3. Wedemeyer H, et al. EASL 2023. Oral #OS-068. 4. Dietz-Fricke C, et al. *JHEP Rep*. 2023;5:100686.

Acknowledgments: We extend our thanks to the patients, their families, and all participating investigators and their corresponding site staff. This study was funded by Gilead Sciences, Inc. Writing and editorial support was provided by Allison Yankey, PhD, of AlphaScientia, a Red Nucleus company, and was funded by Gilead Sciences, Inc.

Disclosures: **PL** reports speaking and teaching fees from and participation in advisory committees or review panels for AbbVie; Aligos Therapeutics; Alnylam Pharmaceuticals; Antios Therapeutics; Arrowhead Pharmaceuticals; Bristol Myers Squibb; Eiger Biopharmaceuticals; Gilead Sciences, Inc.; GSK; Janssen; Merck Sharp & Dohme; MYR Pharmaceuticals; Roche; and Spring Bank Pharmaceuticals. **SA** reports honoraria from AbbVie; Biogen; Gilead Sciences, Inc.; and Merck Sharpe & Dohme; and research grants from AbbVie and Gilead Sciences, Inc. **PB**, **TS**, **AB**, and **SC** report nothing to disclose. **SZ** reports serving on a speakers bureau and/or consulting for AbbVie; Allergan; BioMarin; Gilead Sciences, Inc.; Intercept Pharmaceuticals; Janssen; Merck Sharp & Dohme; Novo Nordisk; Swedish Orphan Biovitrum; and Theratechnologies. **MC** received honoraria from AbbVie; Falk; Gilead Sciences, Inc.; GSK; Janssen-Cilag; Merck Sharp & Dohme; Novartis; Roche; Spring Bank Pharmaceuticals; and Swedish Orphan Biovitrum. **DM**, **ML**, **AHL**, **BLD**, and **GMC** are employees of Gilead Sciences, Inc., and may own stock in Gilead Sciences, Inc. **MRB** reports consulting and serving on a speakers bureau for AbbVie; Eisai–Merck Sharp & Dohme; Gilead Sciences, Inc.; Janssen; and Roche. **HW** reports being a consultant for Abbott; AbbVie; Aligos Therapeutics; Arbutus Biopharma; Boehringer Ingelheim; Bristol Myers Squibb; Dicerna; Gilead Sciences, Inc.; Johnson & Johnson/Janssen-Cilag; Merck/Schering-Plough; MYR Pharmaceuticals; Novartis; Roche; Siemens; Transgene; ViiV Healthcare; and Vir Biotechnology; and reports speaking honoraria from Abbott; AbbVie; Boehringer Ingelheim; Bristol Myers Squibb; Gilead Sciences, Inc.; Johnson & Johnson/Janssen-Cilag; Merck/Schering-Plough; MYR Pharmaceuticals; Novartis; Roche; Siemens; Transgene; and ViiV Healthcare.

Introduction

- Hepatitis delta virus (HDV) represents the most severe form of chronic viral hepatitis and is estimated to affect between 10 and 20 million individuals worldwide[1]
- Bulevirtide (BLV), a novel entry inhibitor of HDV, is fully approved in the European Union, Great Britain, Switzerland, and the Russian Federation at the dose of 2 mg/day for the treatment of chronic hepatitis delta (CHD) with compensated liver disease[2]
- In the MYR301 study, BLV monotherapy (2 or 10 mg) resulted in a virologic response in 76% to 82%, normalisation of alanine aminotransferase (ALT) in 63% to 64%, and a combined response (ALT normalisation + virologic response) in 55% to 56% of patients after 96 weeks[3]
- A recent subanalysis showed that ALT improvement may occur discordantly of virologic response with BLV treatment[4]
- It remains unclear whether histologic improvement correlates with ALT and/or virologic response in patients treated with BLV monotherapy

Objective

- This study aims to evaluate the evolution and correlation of biochemical, virologic, and histologic parameters in patients with CHD treated with BLV monotherapy for 48 weeks

Methods

MYR301 Study Design

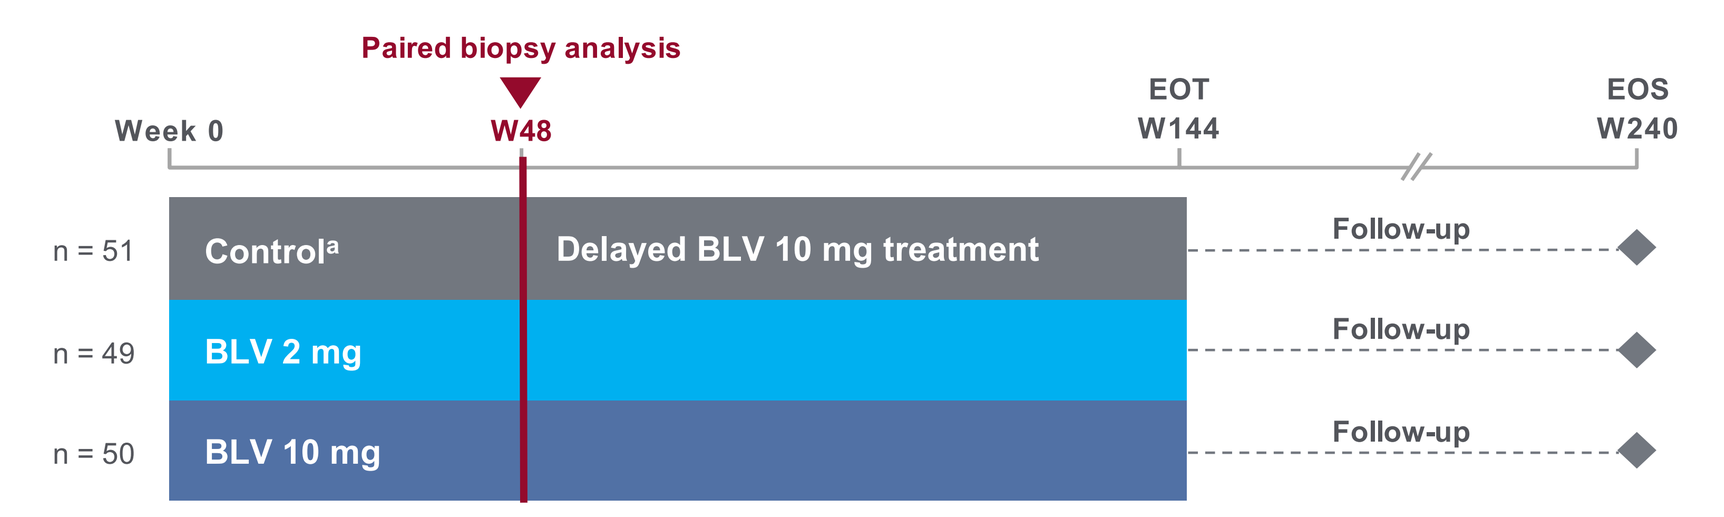
Paired biopsy analysis
Week 0
W48
EOT
W144
EOS
W240
n = 51
Control[a]
Delayed BLV 10 mg treatment
Follow-up
n = 49
BLV 2 mg
Follow-up
n = 50
BLV 10 mg
Follow-up

[a]Patients in the control group for this subanalysis were treated with BLV 10 mg starting at W48. Liver biopsies were performed per protocol at baseline and W48. If baseline liver biopsy samples were not available (were not provided to central laboratory or were considered as nonevaluable by central laboratory), subsequent liver biopsy was not performed. BLV, bulevirtide; EOS, end of study; EOT, end of treatment; W, week.

- This analysis included data up to week (W) 48 from patients in the ongoing randomised Phase 3 Study MYR301 (NCT03852719) who had paired (baseline [BL] and W48) liver biopsies
- At W48, there were 3 study arms: BLV 2 mg, BLV 10 mg, and a delayed-treatment arm that received no anti-HDV therapy to this point (control)
- Viral response groups at W48 were defined as follows:
 - — Virologic response (VR) = HDV RNA decline of ≥2 log_{10} IU/mL from BL or undetectable HDV RNA
 - — Partial response (PR) = HDV RNA decline of ≥1 but <2 log_{10} IU/mL from BL
 - — Nonresponse (NR) = HDV RNA decline of <1 log_{10} IU/mL from BL
- Histologic analysis included:
 - — Histologic parameters: histologic activity index (HAI; 0–18), HAI category (0–4), and Ishak fibrosis score (0–6)
 - — Improvement at W48 was defined as ≥1-point improvement from BL for HAI and Ishak fibrosis score and ≥1 HAI category improvement from BL
 - — Histologic improvement was defined as ≥2-point improvement in HAI with no worsening of fibrosis score
- ALT normalisation was defined at Russian sites as ≤31 U/L for females and ≤41 U/L for males, and at all other sites as ≤34 U/L for females and ≤49 U/L for males
- HDV RNA levels were determined by RT-qPCR using RoboGene® HDV RNA Quantification Kit 2.0 (lower limit of quantification 50 IU/mL, lower limit of detection 6 IU/mL)

Results

Demographics and Baseline Characteristics Based on Virologic Response

	BLV (2 or 10 mg)				
	VR at W48 (n = 45)	PR at W48 (n = 6)	NR at W48 (n = 5)	Control (n = 27)	Total (N = 83)
Age, years, mean (SD)	44 (8.6)	38 (9.2)	46 (10.7)	40 (7.4)	42 (8.6)
Male sex, n (%)	22 (48.9)	4 (66.7)	3 (60.0)	14 (51.9)	43 (51.8)
Race, n (%)					
White	41 (91.1)	6 (100.0)	3 (60.0)	20 (74.1)	70 (84.3)
Asian	3 (6.7)	0	2 (40.0)	7 (25.9)	12 (14.5)
Black or African American	1 (2.2)	0	0	0	1 (1.2)
Cirrhosis present, n (%)	16 (35.6)	1 (16.7)	1 (20.0)	9 (33.3)	27 (32.5)
HBeAg positive, n (%)	6 (13.3)	1 (16.7)	0	3 (11.1)	10 (12.0)
Concomitant NA therapy, n (%)	28 (62.2)	2 (33.3)	3 (60.0)	18 (66.7)	51 (61.4)
Prior IFN therapy, n (%)	33 (73.3)	2 (33.3)	2 (40.0)	16 (59.3)	53 (63.9)
Genotype HDV-1, n (%)[a]	43 (95.6)	6 (100.0)	5 (100.0)	27 (100.0)	81 (97.6)
HDV RNA, log_{10} IU/mL, mean (SD)	5.1 (1.4)	5.2 (1.1)	5.1 (1.8)	5.2 (1.2)	5.1 (1.3)
ALT, U/L, median (Q1, Q3)	92 (63, 134)	96 (90, 175)	138 (133, 146)	77 (54, 116)	92 (63, 136)
LSM, kPa, mean (SD)	12.8 (7.0)	8.1 (2.2)	8.2 (3.9)	12.4 (8.1)	12.1 (7.1)
HAI, mean (SD)	9 (3.4)	8 (3.1)	8 (2.8)	8 (3.3)	8 (3.3)
Ishak fibrosis score, mean (SD)	3 (1.5)	2 (1.6)	2 (0.4)	2 (1.4)	2 (1.4)

[a]In the BLV 10 mg group, 1 patient was HDV genotype 5 and 1 patient was missing HDV genotype data; both patients had VR at W48.
ALT, alanine aminotransferase; BLV, bulevirtide; HAI, histologic activity index; HBeAg, hepatitis B e antigen; HDV, hepatitis delta virus; IFN, interferon; LSM, liver stiffness measurement; NA, nucleos(t)ide analogue; NR, nonresponse; PR, partial response; Q, quartile; VR, virologic response; W, week.

- Overall, 83 patients with paired BL and W48 liver biopsies were included in this subanalysis
 - — BLV treated (n = 56): VR at W48 (n = 45), PR at W48 (n = 6), and NR at W48 (n = 5)
 - — Control (n = 27)

ALT Levels Over Time (BLV Treated + Control)

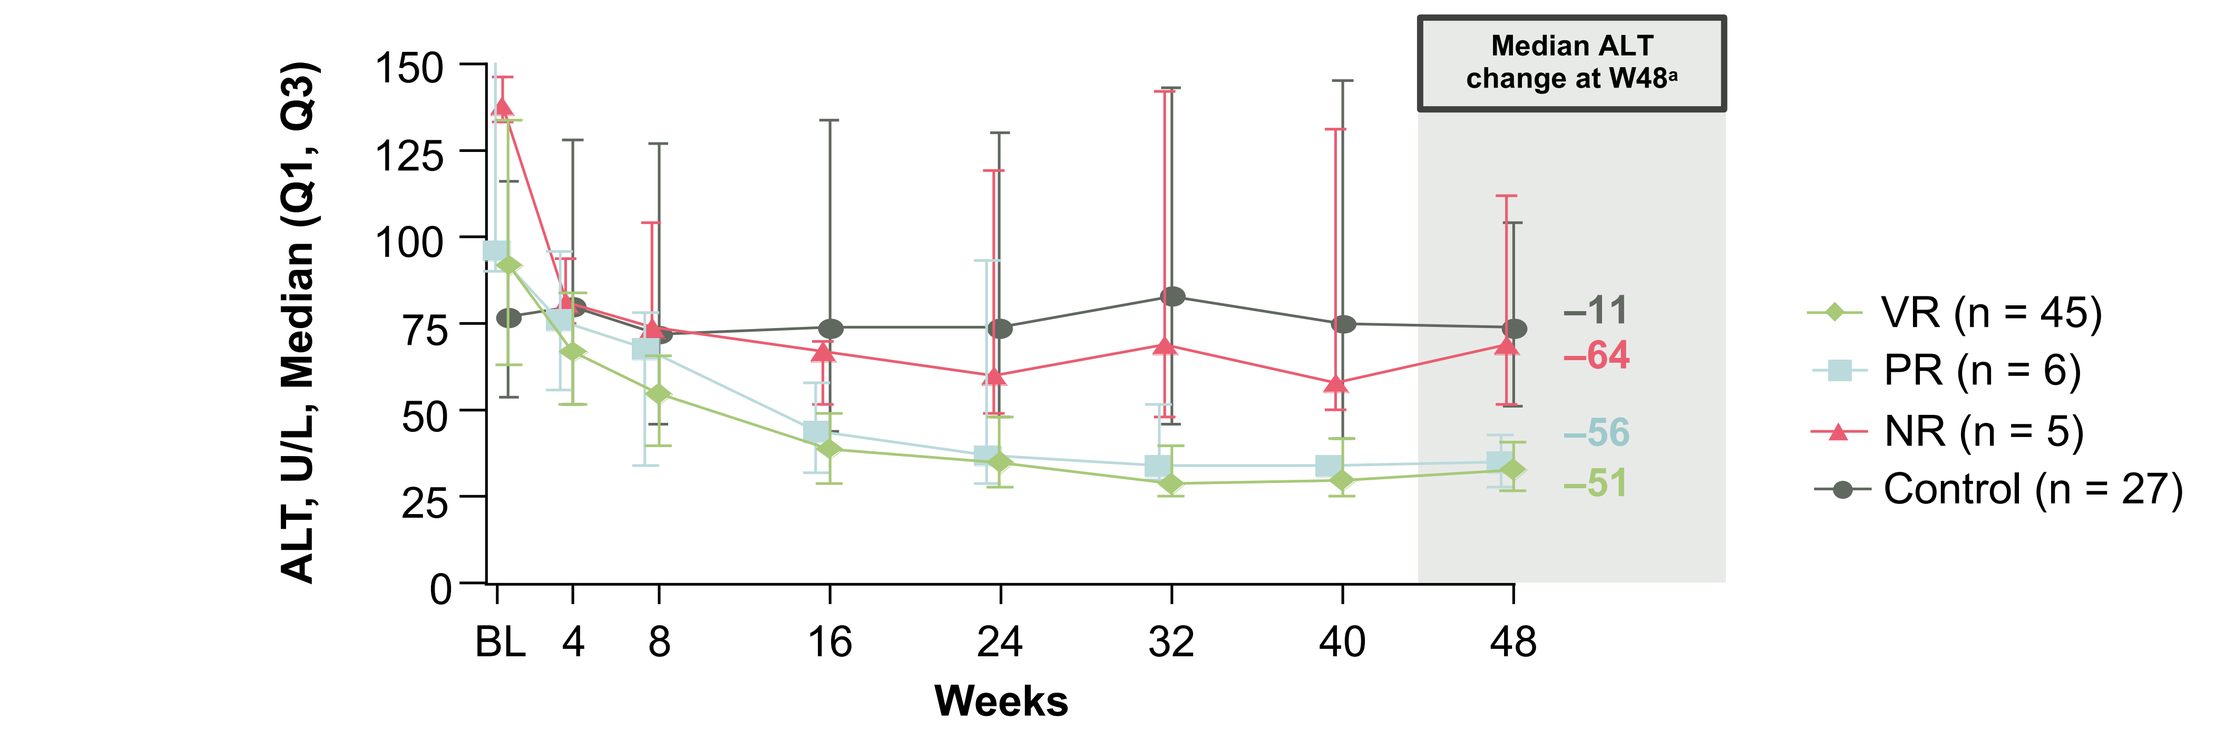
Median ALT change at W48[a]
ALT, U/L, Median (Q1, Q3)
150
125
100
75
50
25
0
BL 4 8 16 24 32 40 48
Weeks
–11
–64
–56
–51
VR (n = 45)
PR (n = 6)
NR (n = 5)
Control (n = 27)

Patients Evaluated	BL	4	8	16	24	32	40	48
VR, n =	45	45	45	45	44	45	44	45
PR, n =	6	6	6	6	6	6	6	6
NR, n =	5	5	5	5	5	5	5	5
Control, n =	27	27	27	25	27	27	26	27

[a]Change at W48 was expressed as median change from BL. Not shown due to scale: ALT Q3 for PR at BL = 175.
ALT, alanine aminotransferase; BL, baseline; BLV, bulevirtide; NR, nonresponse; PR, partial response; Q, quartile; VR, virologic response; W, week.

- Greater median changes in ALT levels from BL were observed in BLV-treated patients compared with the control group at W48, regardless of viral response
 - — Similar median changes in ALT levels were observed across viral response groups in patients treated with BLV

Change From Baseline: HAI by ALT at W48

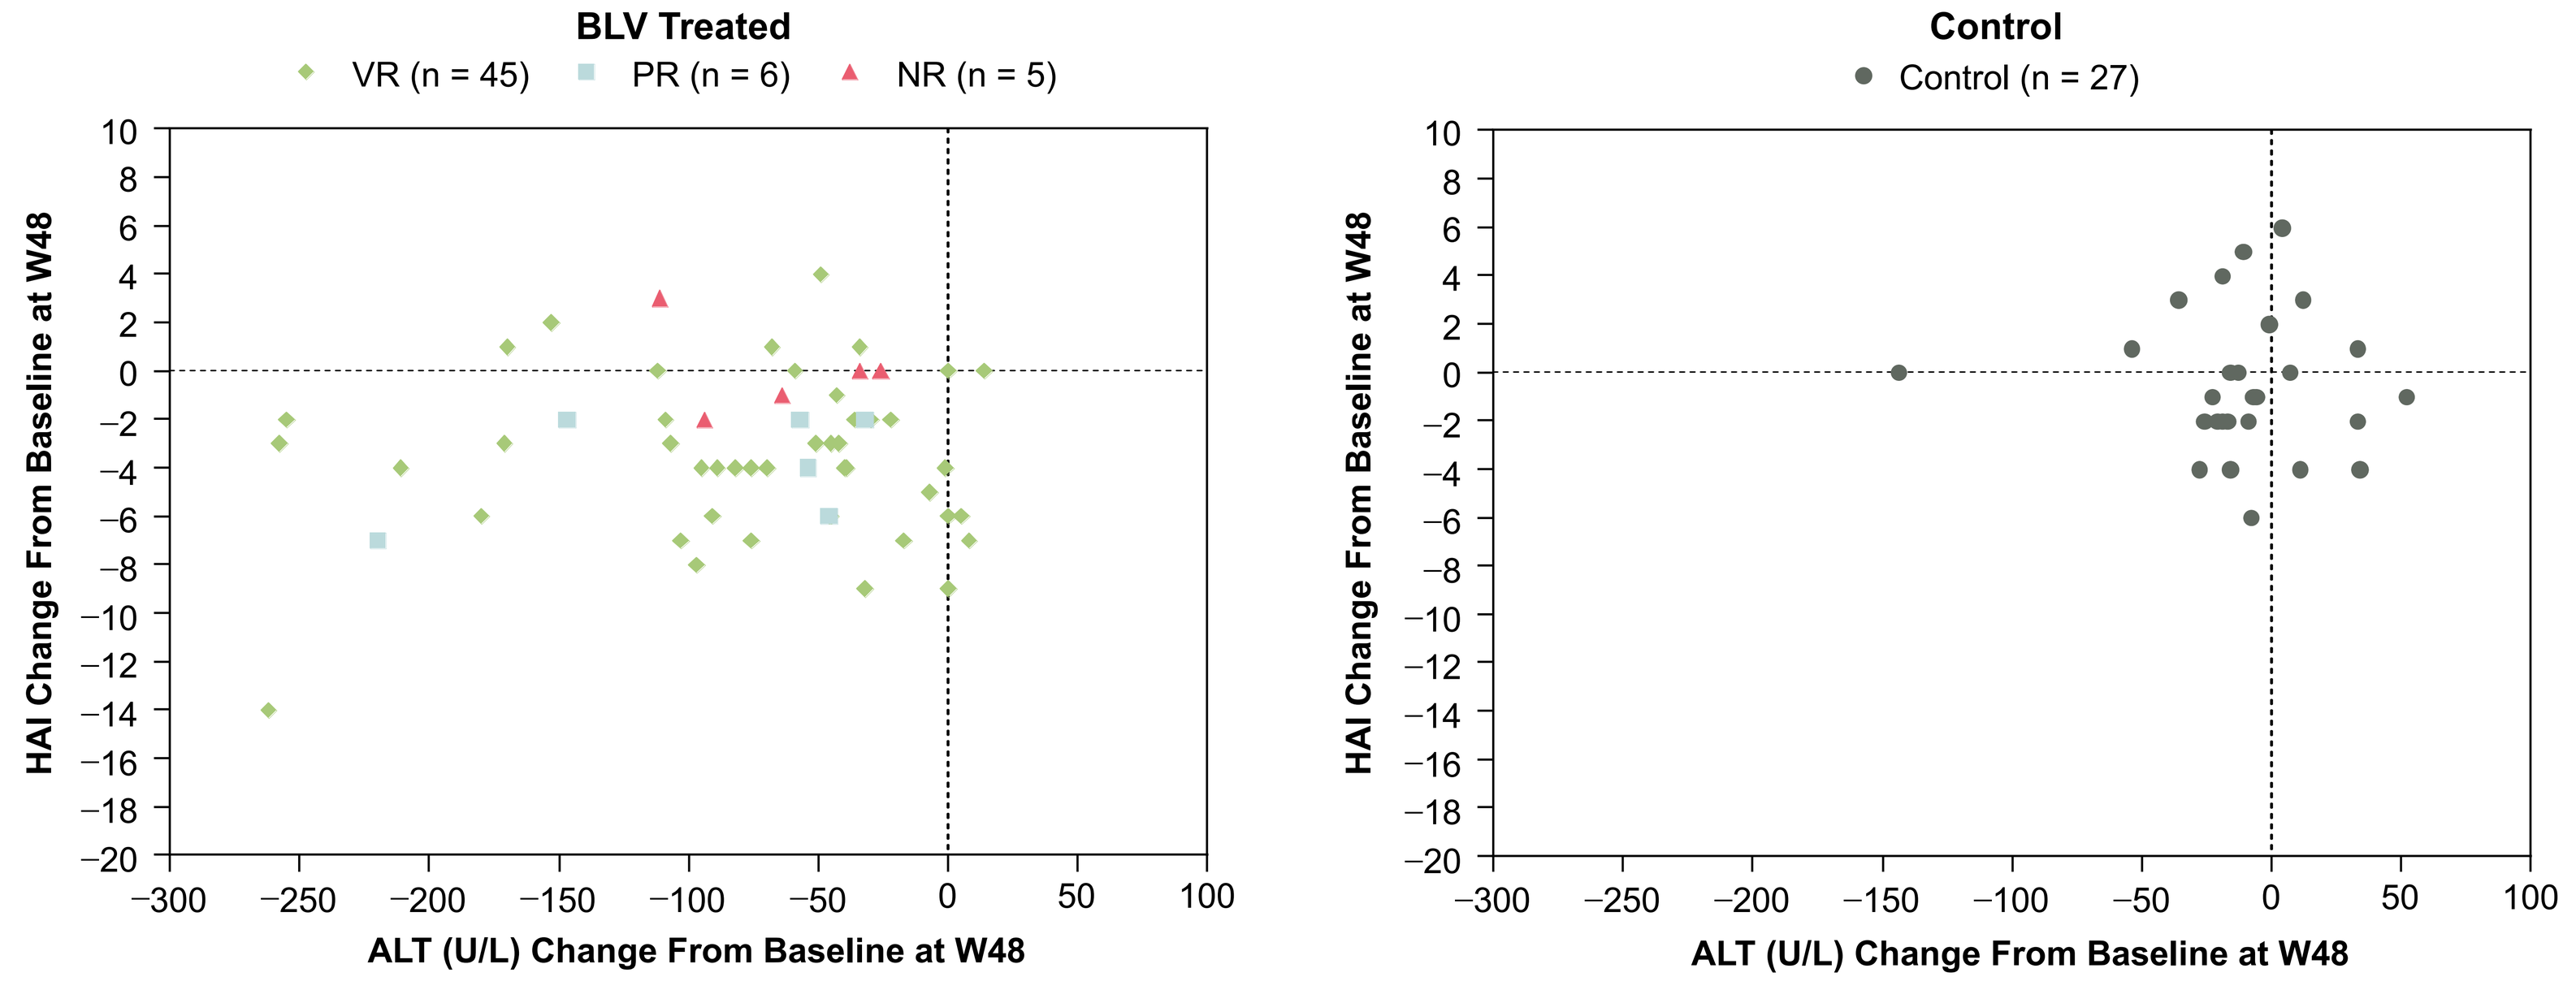
BLV Treated
VR (n = 45)
PR (n = 6)
NR (n = 5)
Control
Control (n = 27)
HAI Change From Baseline at W48
ALT (U/L) Change From Baseline at W48
10
8
6
4
2
0
−2
−4
−6
−8
−10
−12
−14
−16
−18
−20
−300 −250 −200 −150 −100 −50 0 50 100

ALT, alanine aminotransferase; BLV, bulevirtide; HAI, histologic activity index; NR, nonresponse; PR, partial response; VR, virologic response; W, week.

- Decreases in ALT levels and HAI were observed in most patients treated with BLV, irrespective of viral response group
- The degree of improvement in HAI and ALT levels did not necessarily correlate
- No consistent pattern of HAI or ALT level change was observed in the control group

Change in HAI by Viral Response at W48 (BLV Treated + Control)

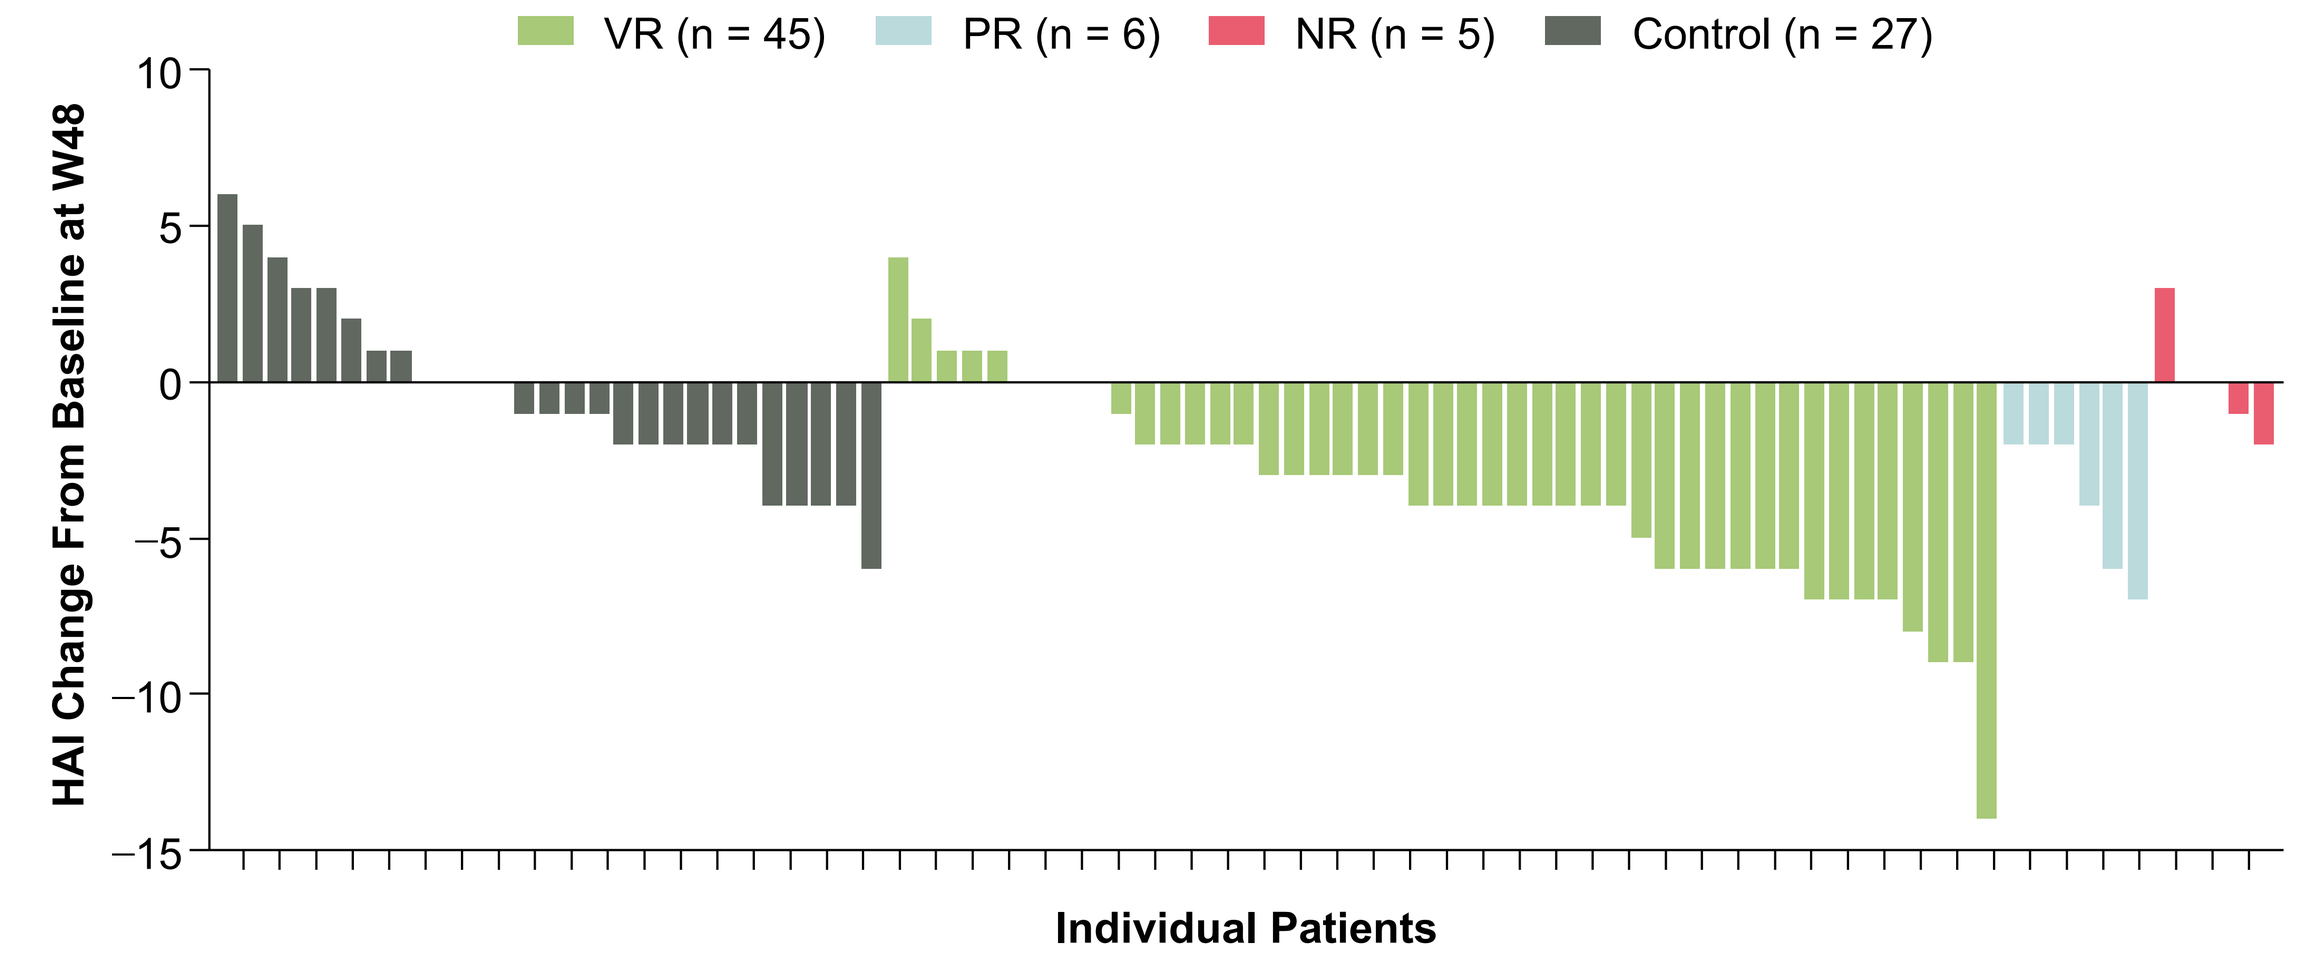
VR (n = 45)
PR (n = 6)
NR (n = 5)
Control (n = 27)
HAI Change From Baseline at W48
10
5
0
−5
−10
−15
Individual Patients

- Improvement in HAI occurred as follows:
 - — VR: 80% (36/45)
 - — PR: 100% (6/6)
 - — NR: 40% (2/5)
 - — Control: 56% (15/27)

Change in HAI by ALT Decline at W48 (BLV Treated)

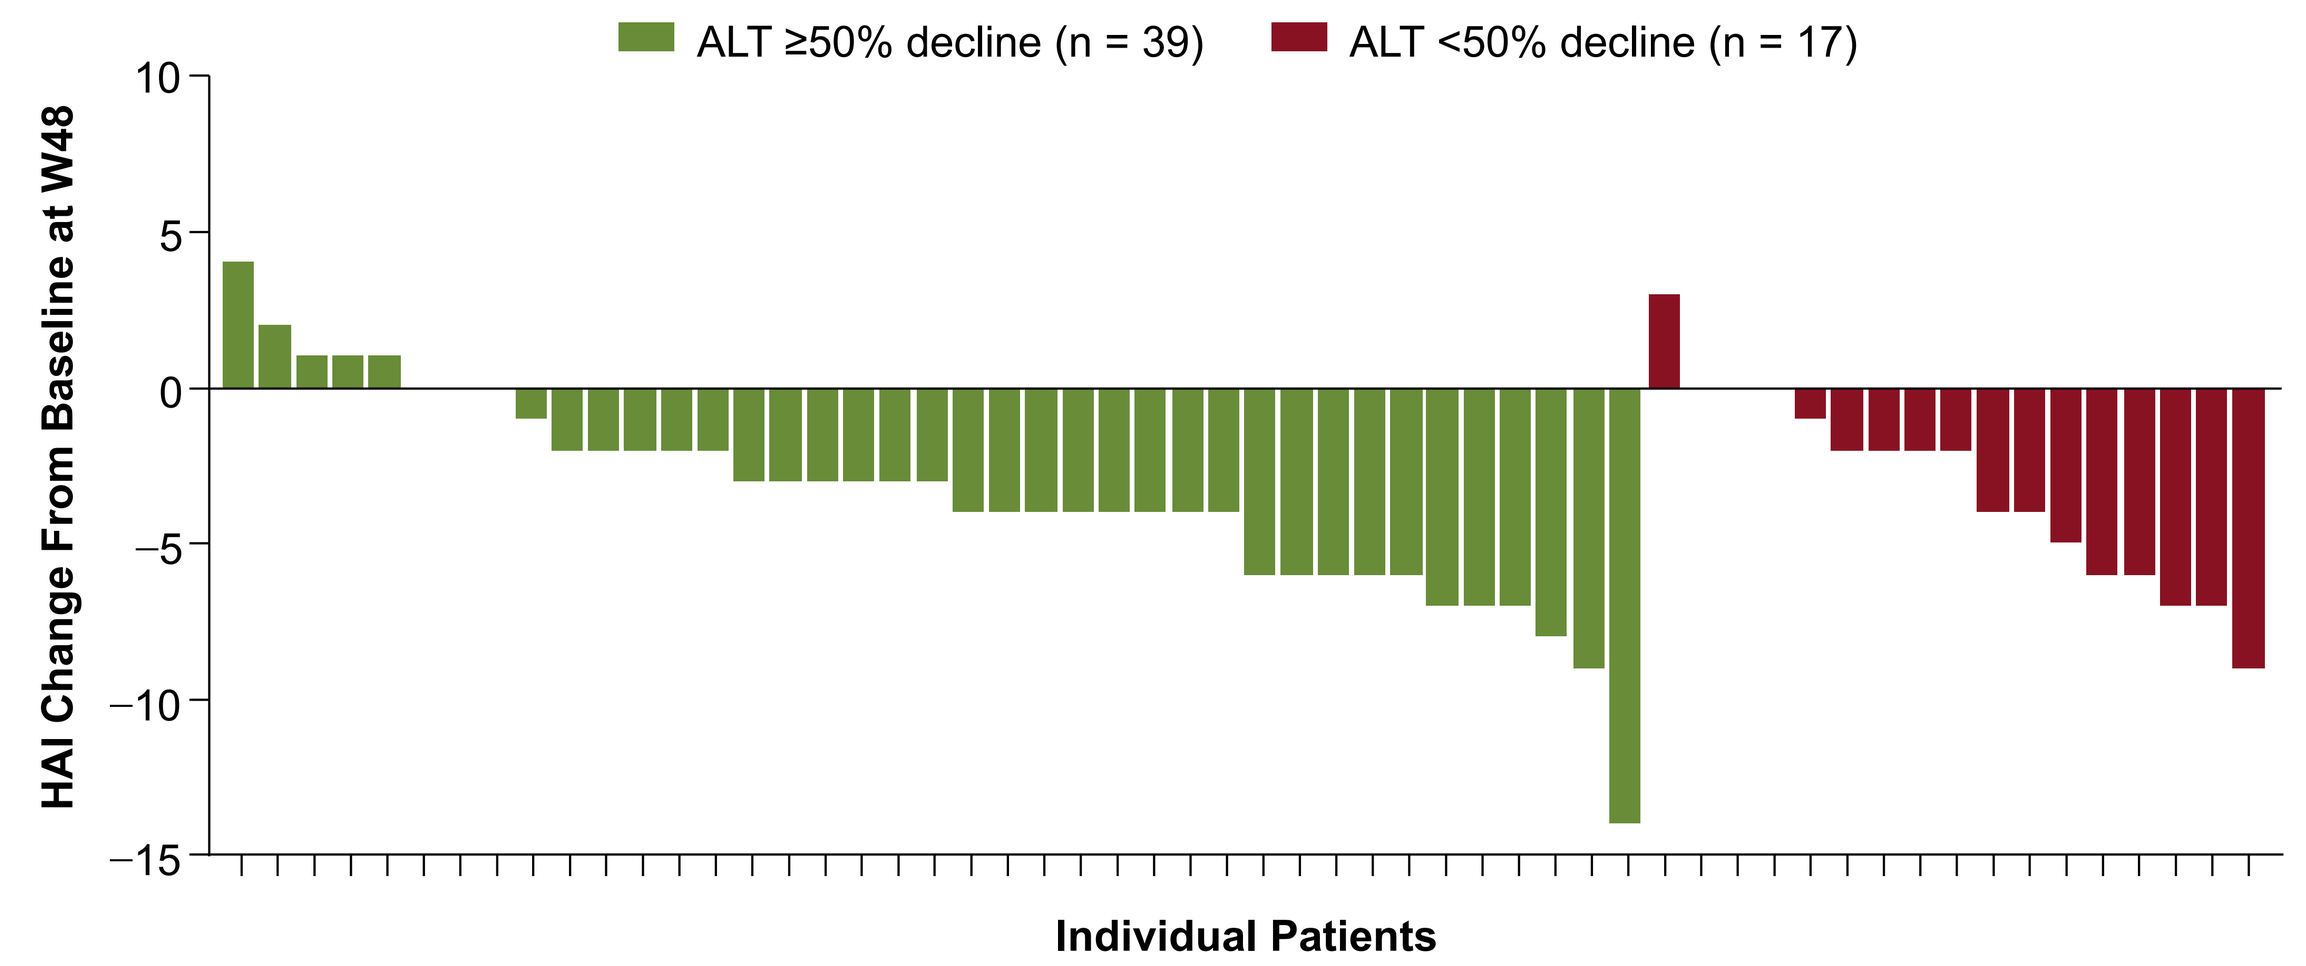
ALT ≥50% decline (n = 39)
ALT <50% decline (n = 17)
HAI Change From Baseline at W48
10
5
0
−5
−10
−15
Individual Patients

- HAI improvement was seen even in patients who had <50% decline in ALT
 - — ALT ≥50% decline: 79% (31/39)
 - — ALT <50% decline: 76% (13/17)

Change in Ishak Fibrosis Score by Viral Response at W48 (BLV Treated + Control)

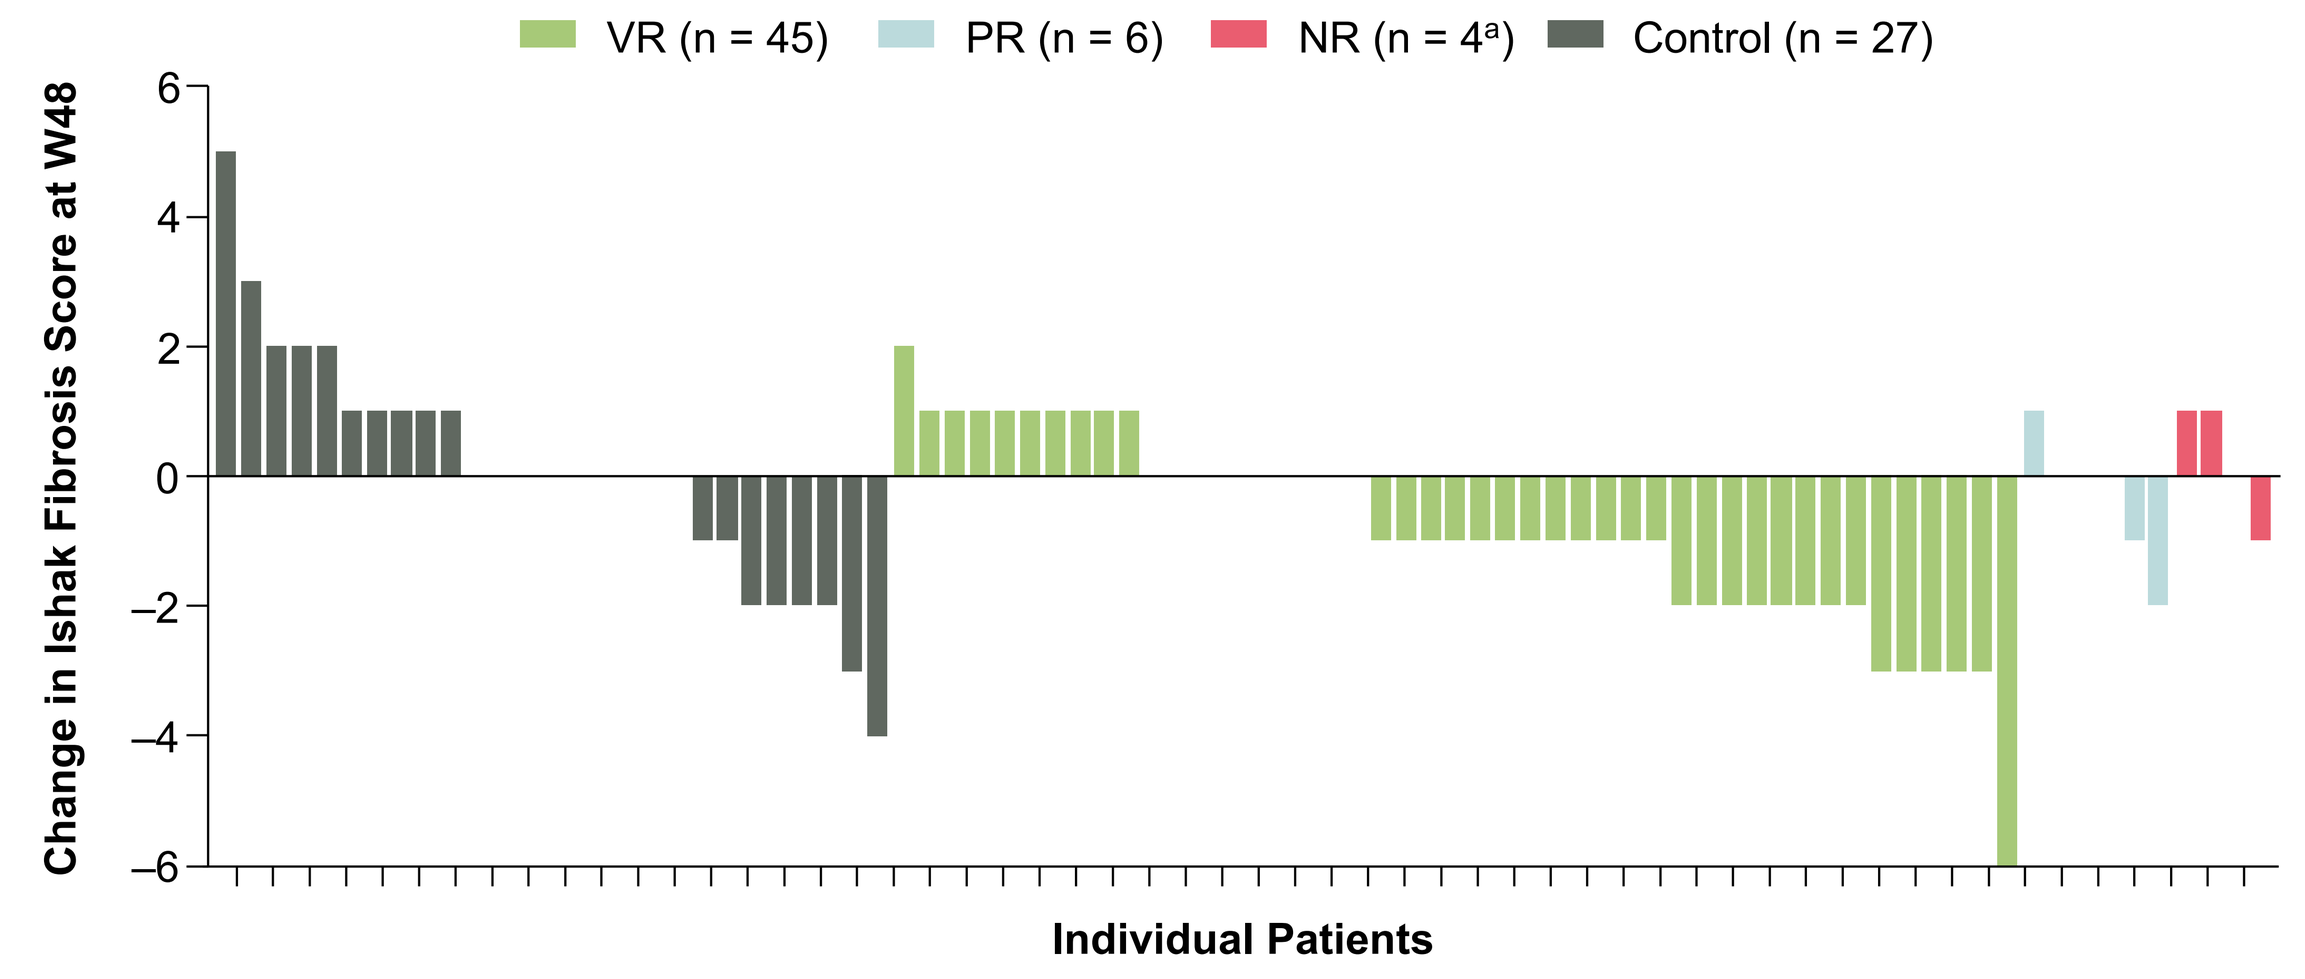
VR (n = 45)
PR (n = 6)
NR (n = 4[a])
Control (n = 27)
Change in Ishak Fibrosis Score at W48
6
4
2
0
−2
−4
−6
Individual Patients

[a]One patient with NR at W48 had no fibrosis score available at W48.
ALT, alanine aminotransferase; BLV, bulevirtide; HAI, histologic activity index; NR, nonresponse; PR, partial response; VR, virologic response; W, week.

- Improvement in Ishak fibrosis score occurred as follows:
 - — VR: 58% (26/45)
 - — PR: 33% (2/6)
 - — NR: 25% (1/4)
 - — Control: 30% (8/27)

Change in HAI Category From Baseline to W48

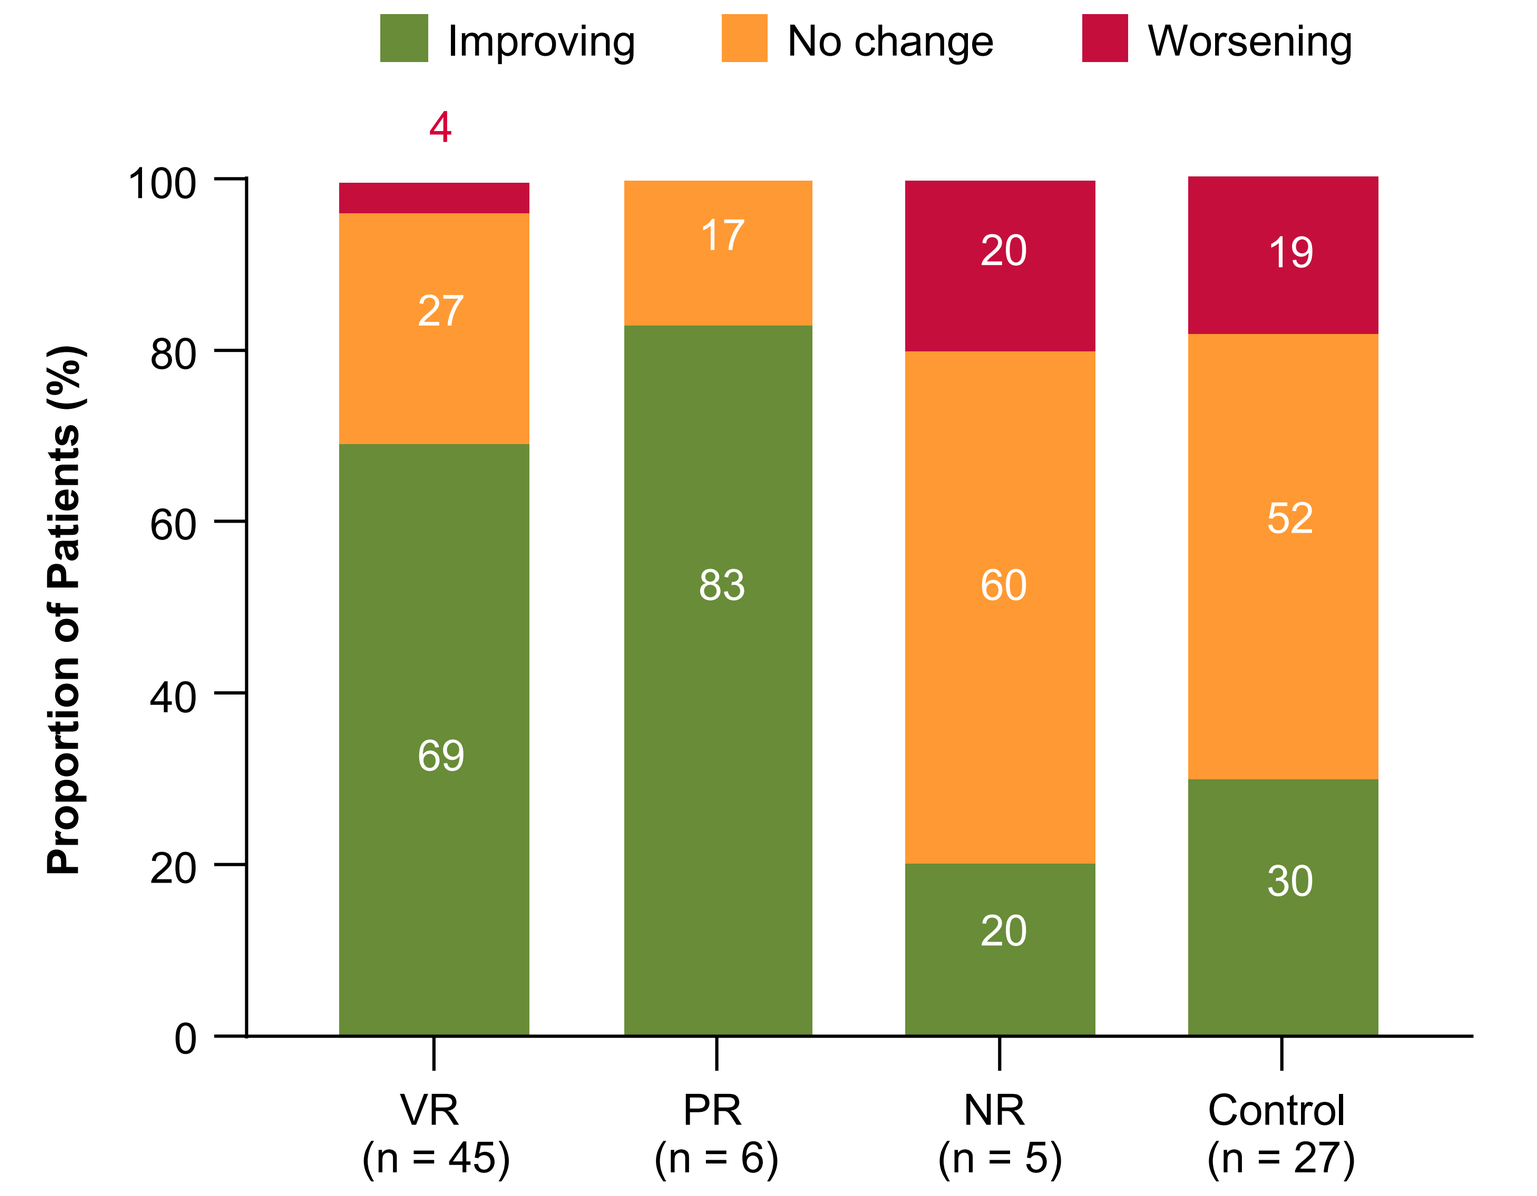
Improving
No change
Worsening
Proportion of Patients (%)
100
80
60
40
20
0
4
27
69
17
83
20
60
20
19
52
30
VR
(n = 45)
PR
(n = 6)
NR
(n = 5)
Control
(n = 27)

HAI category (0–4) based on the HAI (0–18).
HAI, histologic activity index; NR, nonresponse; PR, partial response; VR, virologic response; W, week.

- Improvement in HAI category occurred more frequently in patients with VR and PR

Histologic Improvement From Baseline to W48

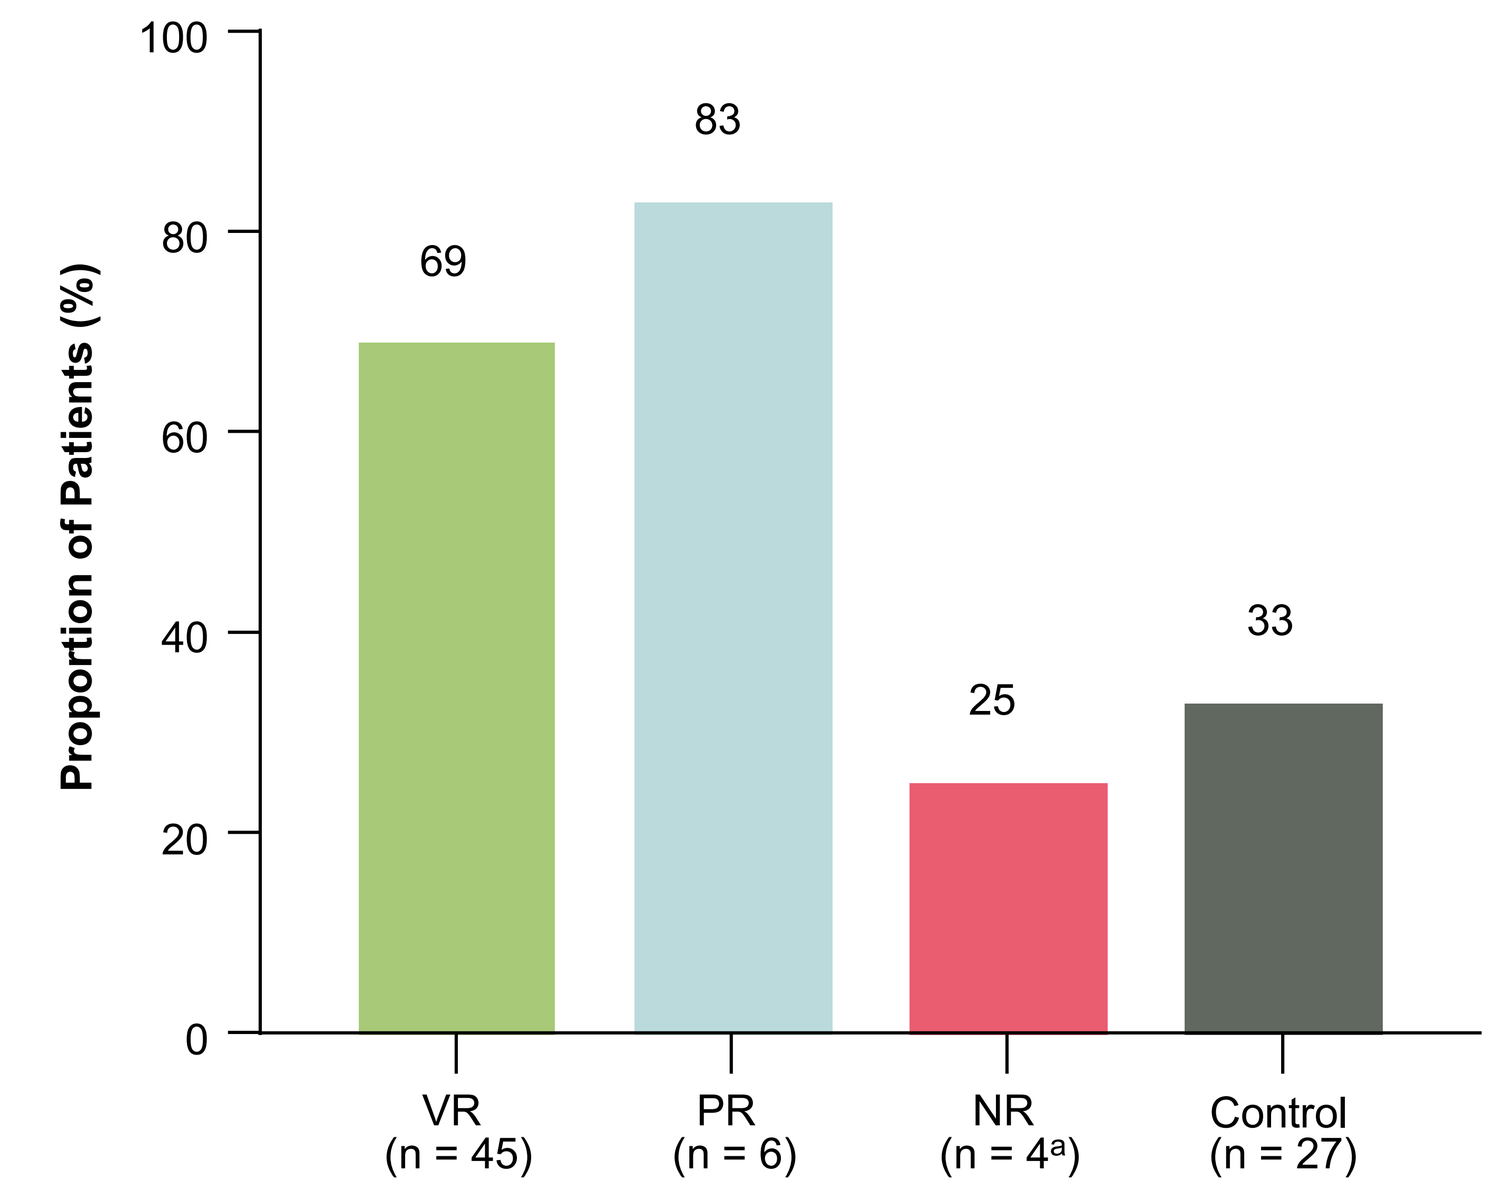
Proportion of Patients (%)
100
80
60
40
20
0
69
83
25
33
VR
(n = 45)
PR
(n = 6)
NR
(n = 4[a])
Control
(n = 27)

[a]One patient with NR at W48 had no fibrosis score available at W48. Histologic improvement was defined as a ≥2-point improvement in HAI with no worsening of Ishak fibrosis score.
HAI, histologic activity index; NR, nonresponse; PR, partial response; VR, virologic response; W, week.

- Histologic improvement occurred more frequently in patients with VR and PR

European Association for the Study of the Liver; 05–08 June 2024; Milan, Italy